DISPOSITION AND METABOLISM OF CICLESONIDE:

PHARMACOKINETICS, METABOLISM, AND EXCRETION IN THE MOUSE, RAT, RABBIT, AND DOG

Zuyu Guo,[1] Stanley R. Howell,[1] Zheming Gu,[2] Kelly Chen,[1] Shashank Rohatagi,[1] Lining Cai,[2] Jinn Wu,[2] John Stuhler,[3] and S. Peter King[1]

[1] Drug Metabolism and Pharmacokinetics, Sanofi-Aventis, Bridgewater, NJ, USA; [2] XenoBiotic Laboratories, Inc., Plainsboro, NJ, USA; [3] DMPK, Quintiles Inc., Kansas City, MO, USA

Abstract

Purpose: Ciclesonide (CIC) is an effective inhaled corticosteroid for the treatment of asthma. CIC is converted to des-isobutyrylciclesonide (des-CIC), the active metabolite, by esterases in the lung where it exerts its anti-inflammatory effects. This study examined the pharmacokinetics (PK), metabolism and excretion of ^{14}C-CIC in the animal species.

Methods: Inhalation dosing of ^{14}C-CIC was not readily feasible in animals, therefore PK and excretion were examined after intravenous (IV) and oral dosing of ^{14}C-CIC. Serum concentrations of CIC and des-CIC were quantitated. Excreta and serum samples were profiled to characterize the metabolic fate of CIC.

Results: In all species, the oral bioavailability of CIC and des-CIC was very low in all species. Both CIC and des-CIC were rapidly cleared and widely distributed. Assuming full conversion of CIC to des-CIC, des-CIC clearance was 7.3, 5.9, 1.6 and 3.5 L/h/kg, respectively, in mice, rats, rabbits and dogs; des-CIC V_d was 75, 43, 13 and 17 L/kg, respectively. The terminal $t_{1/2}$ of CIC was short; the $t_{1/2}$ of des-CIC was longer and ranged from 2.4 to 6.9 h in the four species. CIC was extensively metabolized to yield active des-CIC, which was further metabolized to primarily inactive hydroxylated metabolites-cyclohexane-monohydroxylated des-CIC and B-ring-monohydroxylated
des-CIC. Greater than 90% of IV and oral ^{14}C-CIC doses were recovered in all species; the main elimination route was fecal/biliary while renal was a minor route of elimination.

Conclusion: CIC had low oral bioavailability, high CL, large Vd with fecal/biliary as the major route of elimination. CIC was converted to active des-CIC and further to primarily inactive oxidative metabolites.

Introduction

CIC is a novel and effective non-halogenated glucocorticoid developed for treatment of asthma. CIC has low glucocorticoid receptor affinity. It undergoes conversion to its high affinity active metabolite (des-CIC), via esterases primarily in the target organ, lung. In humans, CIC and des-CIC have very low oral bioavailability (<1%), rapid elimination and a high clearance rate

Mouse, rat, rabbit, and dog are the animal species used for the safety evaluation of CIC. Similar metabolite profiles in these species to those in humans would support the use of these species as appropriate animal species for the safety evaluation of CIC. In the present study, the disposition of ^{14}C-CIC was examined in mice, rats, rabbits and dogs after intravenous (IV) and oral administration to provide information on the absorption, metabolism and excretion of CIC in the species.

Materials and Methods

Male B6C3F1 mice (22-29 g, *n* = 4/dose route/time point for blood sampling, n=6 for mass balance), male Wistar rats (0.25–0.31 kg, *n* = 3/dose route/time point for blood sampling, n=6 for mass balance), female Himalayan rabbits (2.25-2.55 kg, n=6), and male Beagle dogs (7-10 kg, n=6) were given a single IV or oral ^{14}C-CIC dose after an acclimation period. ^{14}C-CIC doses were: mouse, 0.75 mg/kg (IV and oral); rat, 0.5 mg/kg (IV and oral); rabbit, 0.022 mg/kg (IV) and 0.1 mg/kg (oral); dog, 0.1 mg/kg (IV) and 0.5 mg/kg (oral). The IV dose was prepared in a solution of ethanol/1,2-propylene glycol. The oral dose was prepared in a solution of PEG 400/water. Blood samples were collected at various timepoints after dosing; serum or plasma (rabbit) was prepared. Urine and feces were analyzed for radioactivity to assess excretion and mass balance. Selected excreta samples were used for metabolite profiling and identification.

Radioactivity amount or concentrations in various matrices were quantitated by LSC. Serum or plasma concentrations of CIC and des-CIC metabolite were determined by a valiadated LC/MS/MS method. Metabolite radioprofiling was accomplished by using HPLC with fraction collection followed by solid scintillation counting. Mass spectral analyses were performed on a Finnigan LCQ mass spectrometer with an electrospray ionization (ESI) probe, in negative and positive ion modes. PK parameters for CIC, des-CIC metabolite, and radioactivity were determined from the mean (mouse and rat) or individual (rabbit and dog) concentration versus time data. Parameter values were determined by noncompartmental methods using WinNonlin™.

Results

Pharmacokinetics: In mice and rats studies where plasma concentration-time profiles of CIC after IV dosing permitted PK analysis, the CL values of CIC were 8.6 and 7.4 L/h/kg, respectively in mice and rats; the apparent Vdss of CIC were 5.1 and 4.2 L/kg, respectively in mice and rats. The CL of CIC in both mice and rats exceeded the hepatic blood flow rate and the Vdss of CIC in both mice and rats exceeded the volume of total body water. Assuming, full conversion of CIC to des-CIC, the calculated CL of des-CIC metabolite was 7.3 L/h/kg, 5.9 L/h/kg, 1.6 L/h/kg 3.45 L/h/kg in mouse, rat, rabbit and dogs, respectively. The calculated Vdarea for des-CIC was 75.4 L/kg, 43.0 L/kg, 13.8 L/kg, 16.6 L/kg in mice, rats, rabbits and dogs, respectively. The CL of des-CIC in all species exceeded or was similar to the hepatic blood flow rate and the Vdarea of des-CIC in all species exceeded the volume of total body water. After both IV and oral dosing, CIC was rapidly converted to des-CIC. However, in all species, the oral exposures of CIC and des-CIC combined was ~6% or less relative to the exposures after IV due to extensive first-pass metabolism of both compounds.

Excretion of Radioactivity: In the mouse, rat, rabbit, and dog, over 90% recovery of the radioactive dose was achieved after both IV and oral dosing. With majority of the recovered radioactive dose excreted in the first 48 hrs. The radioactive dose excreted in feces ranged from 70.5% to 86.9%.

Metabolite Profiles and Identification: des-CIC and hippuric acid were found to be the main metabolites of ^{14}C-CIC in mice, rats, rabbits and dogs. B-ring–monohydroxylated des-CIC and cyclohexane-monohydroxylated des-CIC isomers were also detected in dogs, but not the other species. There were numerous other metabolites that could not be identified due to weak mass spectra signals.

TABLE 1 - Serum PK parameter values for CIC, des-CIC and radioactivity in mouse, rat, rabbit (plasma), and dog given a single IV or oral suspension dose of ^{14}C-CIC

Parameter	CIC		Des-CIC		Radioactivity	
Mouse	IV	Oral	IV	Oral	IV	Oral
(N=4/timepoint)	0.75 mg/kg	0.75 mg/kg	0.75 mg/kg	0.75 mg/kg	0.75 mg/kg	0.75 mg/kg
C_{max} (ng/mL)	198 ± 90.3[a]	ND	109 ± 27.7	0.33 ± 0.18	596 ± 91.1[a]	40.54 ± 6.25
T_{max} (h)	NA	ND	0.083	0.50	NA	0.25
$AUC_{0-?}$ (ng.h/mL)	82.04	ND	99.61	1.39[b]	966	201
$t_{1/2}$ (h)	0.97	ND	6.94	2.54[b]	7.34[b]	3.37
CL (L/h/kg)	8.6	NA	7.3*	NA	NA	NA
Vdss (L/kg)	5.13	NA	75.4**	NA	NA	NA
Rat	IV	Oral	IV	Oral	IV	Oral
(n=3/timepoint)	0.50 mg/kg	0.50 mg/kg	0.50 mg/kg	0.50 mg/kg	0.50 mg/kg	0.50 mg/kg
Cmax (ng/mL)	158 ± 37.7[a]	ND	73.22 ± 8.61	0.289 ± 0.426	400 ± 73.3[a]	18.99 ± 2.61
Tmax (h)	NA	ND	0.083	0.083	NA	4
$AUC_{0-?}$ (ng.h/mL)	63.80	ND	84.71	0.805	653	226
$t_{1/2}$ (h)	1.02	ND	5.05	3.35	21.33	18.15
CL (L/h/kg)	7.42	NA	5.9*	NA	NA	NA
Vdss (L/kg)	4.17	NA	43.0**	NA	NA	NA
Rabbit	IV	Oral	IV	Oral	IV	Oral
(n=6)	0.022 mg/kg	0.10 mg/kg	0.022 mg/kg	0.10 mg/kg	0.022 mg/kg	0.10 mg/kg
Cmax (ng/mL)	0.637 ± 0.238[a]	ND	8.25 ± 2.17	ND	11.65 ± 1.58[a]	4.65 ± 1.26
Tmax (h)	NA	ND	0.083 ± 0.00	ND	NA	0.8 ± 0.3
$AUC_{0-?}$ (ng.h/mL)	ND	ND	13.50 ± 3.02	ND	26.30 ± 6.55	23.23 ± 6.19
$t_{1/2}$ (h)	ND	ND	2.35 ± 0.65	ND	3.72 ± 2.23	3.78 ± 1.74
CL (L/h/kg)	ND	NA	1.48*	NA	NA	NA
Vdss (L/kg)	ND	NA	5.0**	NA	NA	NA
Dog	IV	Oral	IV	Oral	IV	Oral
(n=6)	0.10 mg/kg	0.50 mg/kg	0.10 mg/kg	0.50 mg/kg	0.10 mg/kg	0.50 mg/kg
Cmax (ng/mL)	121 ± 10.4[a]	3.97 ± 2.23	17.58 ± 2.40	3.76 ± 2.04	142 ± 11.5[a]	94.08 ± 27.52
Tmax (h)	NA	0.42 ± 0.13	0.38 ± 0.14	0.79 ± 0.64	NA	1.0 ± 0.5
$AUC_{0-\infty}$ (ng.h/mL)	ND	ND	28.86 ± 3.13	8.42 ± 2.08	405 ± 109	657 ± 111
$t_{1/2}$ (h)	ND	ND	3.32 ± 0.60	3.31 ± 0.74	39.15 ± 8.47	13.49 ± 1.16
CL (L/h/kg)	ND	NA	3.47*	NA	NA	NA
Vdss (L/kg)	ND	NA	16.6**	NA	NA	NA

TABLE 2 - Mean (±SD, n=6) cumulative (0-168 h) excretion of radioactivity after a single IV or oral dose of ^{14}C-CIC in the mouse, rat, rabbit, and dog

Species	% of radioactive dose excreted: Urine	Feces	Cage rinse	Total
Mouse				
IV, 0.75 mg/kg	9.23 ± 2.60	76.3 ± 2.30	5.18 ± 2.36	90.7 ± 2.10
Oral, 0.75 mg/kg	9.71 ± 1.93	70.5 ± 7.40	13.1 ± 6.90	93.3 ± 2.20
Rat				
IV, 0.50 mg/kg	7.41 ± 0.99	84.7 ± 1.20	1.12 ± 0.29	93.2 ± 0.90
Oral, 0.50 mg/kg	8.44 ± 1.35	61.0 ± 7.10	23.9 ± 7.70	93.3 ± 0.70
Rabbit				
IV, 0.022 mg/kg	8.84 ± 1.36	82.9 ± 2.70	1.26 ± 0.87	93.0 ± 1.70
Oral, 0.10 mg/kg	6.91 ± 0.43	86.9 ± 5.20	0.76 ± 0.24	94.5 ± 5.10
Dog				
IV, 0.10 mg/kg	10.0 ± 1.90	78.8 ± 5.60	1.69 ± 1.31	90.5 ± 2.60
Oral, 0.50 mg/kg	8.97 ± 1.10	83.3 ± 1.70	2.00 ± 0.78	94.3 ± 1.50

TABLE 3 - Mass spectral data for molecular ions and the characteristic fragments of CIC and major metabolites generated in vivo in animal species

ID	Structure or Proposed Structure	Characteristic Mass Ions (m/z)	Matrices
CIC	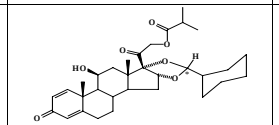	541 ([M+H]$^+$), 341, 323	Mouse_S&F Rat_S&F Rabbit_P&F Dog_S&F
des-CIC	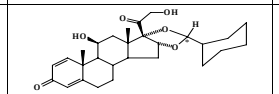	471 ([M+H]$^+$), 341, 323	Mouse_S&F Rat_S&F Rabbit_P&F Dog_S&F
Hippuric Acid	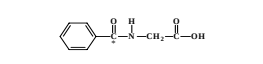	178 ([M-H]$^-$), 134	Mouse_S&U Rat_S&U Rabbit_P&U Dog_S&U
Cyclohexane-hydroxylated des-CIC	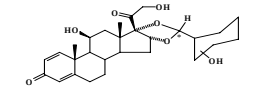	489 ([M+H]$^+$), 341, 323	Rat_S&U Rabbit_P&F Dog_S&F
B-ring-hydroxylated des-CIC	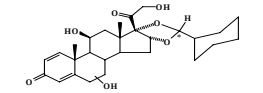	489 ([M+H]$^+$), 357, 323	Dog_S&F
Keto derivative of des-CIC	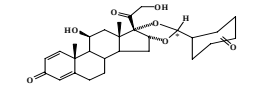	487 ([M+H]$^+$), 341, 323	Rat_S

F: Feces, P: plasma, S: serum, and U: urine.

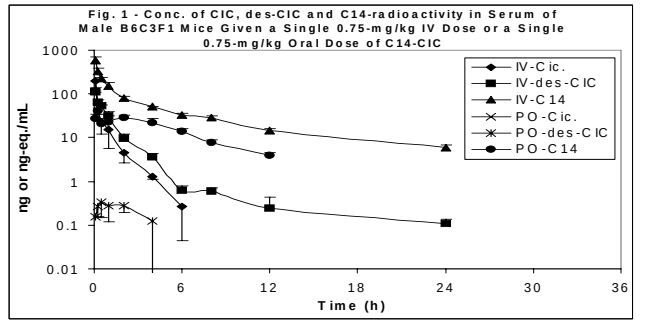
1000
100
10
1
0.1
0.01
ng or ng-eq./mL
0
6
12
18
24
30
36
Time (h)
IV-Cic.
IV-des-CIC
IV-C14
PO-Cic.
PO-des-CIC
PO-C14
Fig. 1 - Conc. of CIC, des-CIC and C14-radioactivity in Serum of Male B6C3F1 Mice Given a Single 0.75-mg/kg IV Dose or a Single 0.75-mg/kg Oral Dose of C14-CIC

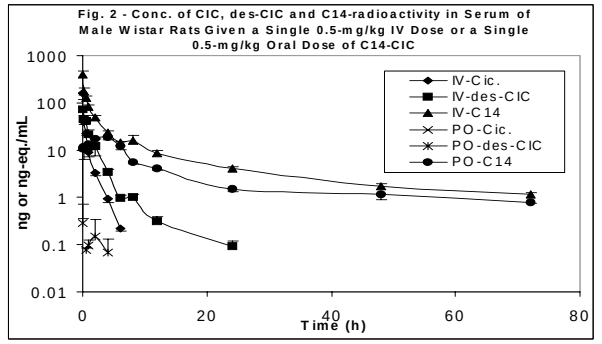
1000
100
10
1
0.1
0.01
ng or ng-eq./mL
0
20
40
60
80
Time (h)
IV-Cic.
IV-des-CIC
IV-C14
PO-Cic.
PO-des-CIC
PO-C14
Fig. 2 - Conc. of CIC, des-CIC and C14-radioactivity in Serum of Male Wistar Rats Given a Single 0.5-mg/kg IV Dose or a Single 0.5-mg/kg Oral Dose of C14-CIC

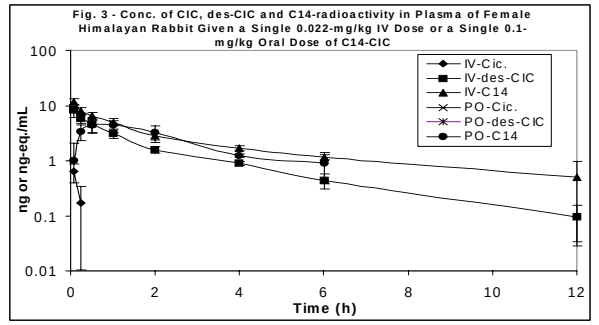
100
10
1
0.1
0.01
ng or ng-eq./mL
0
2
4
6
8
10
12
Time (h)
IV-Cic.
IV-des-CIC
IV-C14
PO-Cic.
PO-des-CIC
PO-C14
Fig. 3 - Conc. of CIC, des-CIC and C14-radioactivity in Plasma of Female Himalayan Rabbit Given a Single 0.022-mg/kg IV Dose or a Single 0.1-mg/kg Oral Dose of C14-CIC

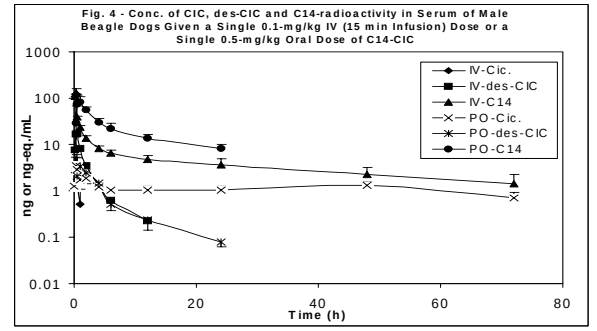
1000
100
10
1
0.1
0.01
ng or ng-eq./mL
0
20
40
60
80
Time (h)
IV-Cic.
IV-des-CIC
IV-C14
PO-Cic.
PO-des-CIC
PO-C14
Fig. 4 - Conc. of CIC, des-CIC and C14-radioactivity in Serum of Male Beagle Dogs Given a Single 0.1-mg/kg IV (15 min Infusion) Dose or a Single 0.5-mg/kg Oral Dose of C14-CIC

Conclusions

- **The PK characteristics of CIC in animal species include high CL, large Vd, and low oral bioavailability.**
- **The very low oral bioavailability (~6% or less) of the active compound species suggests minimal systemic exposure following inhalation of CIC, and is consistent with the low incidence of systemic adverse effects for CIC observed in clinical studies.**
- **Fecal excretion of ^{14}C-CIC-related radioactivity predominated in mouse, rat, rabbit and dog, indicating that biliary excretion and/or gastrointestinal secretion was the major elimination route for CIC-derived radioactivity.**
- **CIC was extensively metabolized to yield active des-CIC, which was further metabolized to primarily inactive hydroxylated metabolites, i.e., cyclohexane-monohydroxylated des-CIC and B-ring-monohydroxylated des-CIC.**
- **Comparisons of both *in vitro* and *in vivo* metabolite profiles between mice, rats, rabbits, and dogs with those from humans indicated that the similar metabolic pathways for CIC were active in humans and in the animal species used for the safety evaluation of CIC.**